71 P

FIN-EGFRprint: A retrospective observational study to investigate treatments and outcomes in patients with epidermal growth factor receptor-mutated (EGFRm) advanced Non-Small Cell Lung Cancer (aNSCLC) in Finland

Aija Knuuttila[1], Lalli Nurmi[2], Petri Vänni[2], Joanne Edwards[3], Monica Ekblom[4], Irene Luccarini[5], Maria Silvoniemi[6]

1Helsinki University Central Hospital, 2Nordic Healthcare Group, Finland, 3Janssen-Cilag Ltd, High Wycombe, United Kingdom, a Johnson & Johnson company, 4Jansen-Cilag Oy, Espoo, Finland, a Johnson & Johnson company, 5Janssen-Cilag SPA, Milan, Italy, a Johnson & Johnson company, 6Turku University Central Hospital

Key Takeaway

This retrospective study in Finland showed that RW median TTNT and OS have improved over time with the transition from 1st- to 2nd- and 3rd-generation TKIs. However, outcomes remain poor, highlighting the need for treatment innovation and new therapies.

Conclusions

Median TTNT significantly increased over time, from 9.7 months (2010–2016) to 21.6 months (2020–2023), with the longest TTNT observed in patients receiving 3rd-gen TKIs (24.3 months).

Median OS improved from 19.1 months (2010–2016) to 29.3 months (2020–2023), with the longest OS observed in patients receiving 3rd-gen TKIs (31.1 months).

While treatment outcomes of NSCLC patients have improved, RW survival with TKIs remains lower than clinical trials results emphasizing the unmet need.

Please scan QR code

Poster

Congress Hub Presentation URL Link
https://jsh.epg-digital.com/Oncology/ELCC2025/Amivantamab/Knuuttila-Advanced

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Acknowledgments

This study was funded by Janssen Pharmaceutica NV. Writing assistance was provided by Eija Heikkilä of NHG and was funded by Janssen Pharmaceutica NV. The study utilized data from hospital data lakes and national registers from 2010 to 2023 in Finland.

Disclosures

Lalli Nurmi received consulting fees paid to their employer by Johnson & Johnson company.

Introduction

- Treatment for EGFRm aNSCLC has substantially evolved over the past decade, thanks to the discovery of activating EGFR mutations, mainly exon 19 deletions and exon 21 L858R substitutions, also defined as common EGFR mutations (cEGFRm)[1-2].
- EGFR tyrosine kinase inhibitors (TKIs) were introduced as first-line (1L) treatment for cEGFRm aNSCLC and have developed over time in 3 generations of drugs, with 3rd gen TKIs currently representing the standard of care (SoC)[3-4].
- Not all TKIs have demonstrated OS benefit within the time frame of clinical trials, therefore long-term real-world (RW) studies are needed to show the evolution of cEGFRm aNSCLC care and how different generations of TKIs have improved patient outcomes.

Methods

Study design, cohort and timelines

- This retrospective observational study utilized national registry data and hospital data lakes from two Finnish university hospitals from 2010 to 2023, covering 55% of NSCLC cases in Finland.
- The study cohort included aNSCLC patients with cEGFR mutations treated in their 1L setting. The aNSCLC patients were identified based on the advanced treatments including TKIs or chemotherapy (CT).

Data-analysis and statistical methods

- Kaplan-Meier survival analysis was used to estimate the probability of OS and TTNT from start of the 1L treatment, and Cox regression analysis was used to compare outcomes between groups.
- The outcomes were analyzed over three time periods based on the introduction in Finland of 1st-gen TKIs (1.1.2010–31.12.2016), 2nd –gen TKIs (1.1.2017–30.9.2020), and 3rd-gen TKIs (1.10.2020–30.9.2023).

Figure 1: Flowchart of study population inclusion

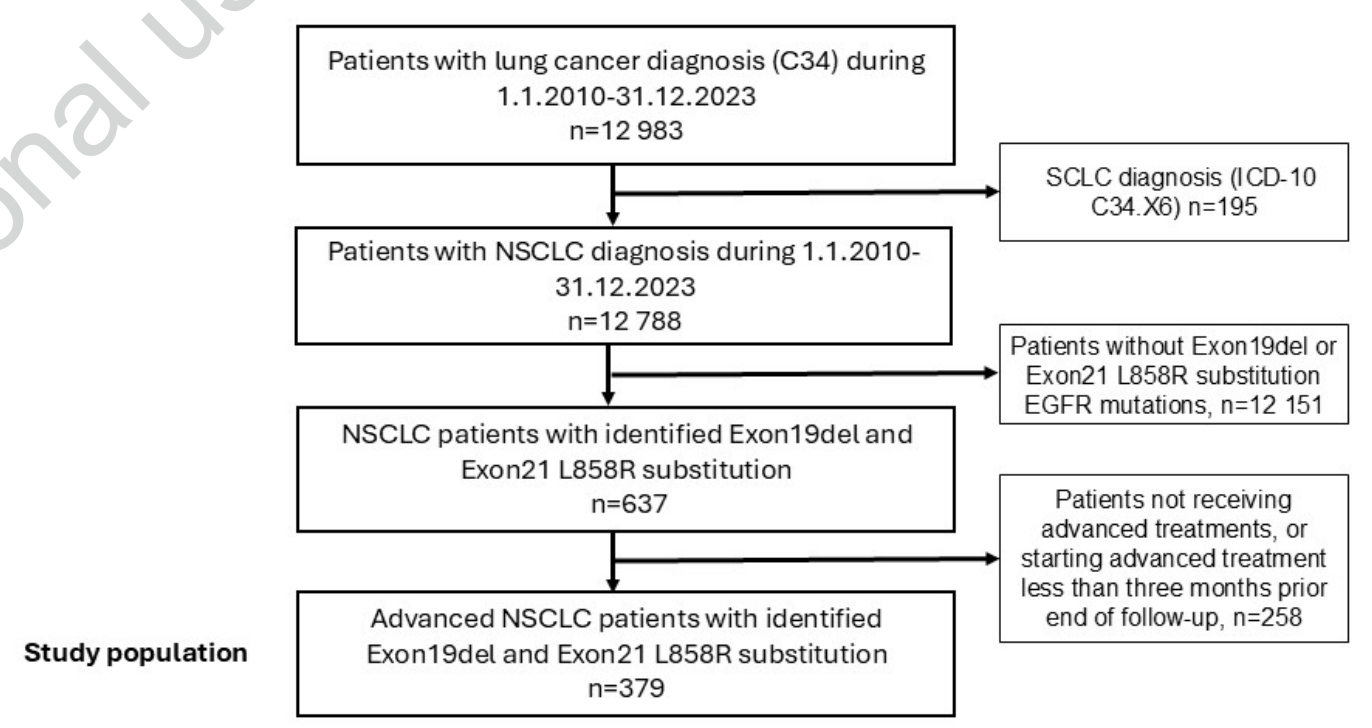
Patients with lung cancer diagnosis (C34) during 1.1.2010-31.12.2023 n=12 983
SCLC diagnosis (ICD-10 C34.X6) n=195
Patients with NSCLC diagnosis during 1.1.2010-31.12.2023 n=12 788
Patients without Exon19del or Exon21 L858R substitution EGFR mutations, n=12 151
NSCLC patients with identified Exon19del and Exon21 L858R substitution n=637
Patients not receiving advanced treatments, or starting advanced treatment less than three months prior end of follow-up, n=258
Study population
Advanced NSCLC patients with identified Exon19del and Exon21 L858R substitution n=379

Results

Study population and patient characteristics (Table 1)

- A total of 379 patients were identified as cEGFRm aNSCLC patients between January 1, 2010, and September 30, 2023 (Figure 1).
- The mean age of the cohort was 69.5 years, with 54% harboring an exon 19 deletion. Most patients (85%) were diagnosed at Stage IV, and 42% presented with metastases to brain, liver, or bone (Table 1).
- The use of CT as 1L therapy declined from 32% (n=41) in 2010–2016 to 6% (n=7) in 2020–2023. 3rd-gen TKI was only used during 2020–2023, when 80% (n=99) of patients received them as 1L treatment.

Table 1: Patient characteristics

Characteristics	All patients n=379	Time Periods 2010-2016 n=129	2017-2020 n=127	2020-2023 n=123
Female, n (%)	260 (69%)	86 (67%)	87 (69%)	87 (71%)
Mean age (SD)	69.5 (11)	67.4 (11)	70.4 (11)	70.6 (11)
Age group, n (%)				
<65 years	117 (31%)	46 (36%)	32 (25%)	39 (32%)
65-75 years	145 (38%)	53 (41%)	55 (43%)	37 (30%)
>75 years	117 (31%)	30 (23%)	40 (31%)	47 (38%)
Stage, n (%)				
Stage IIIb	55 (15%)	28 (22%)	13 (10%)	14 (11%)
Stage IV	324 (85%)	101 (78%)	114 (90%)	109 (89%)
EGFR mutation , n (%)				
Exon 19 deletion	205 (54%)	76 (59%)	68 (54%)	61 (50%)
L858R mutation in exon21	174 (46%)	53 (41%)	59 (46%)	62 (50%)
ECOG grade, n (%)				
0	21 (6%)	<5	#	9 (7%)
1	67 (18%)	20 (16%)	29 (23%)	18 (15%)
≥2	15 (4%)	<5	<5	8 (7%)
Missing	276 (73%)	103 (80%)	85 (67%)	88 (72%)
Metastasis location, n (%)				
Brain	62 (16%)	14 (11%)	28 (22%)	20 (16%)
Liver	51 (13%)	12 (9%)	19 (15%)	20 (16%)
Bone	112 (30%)	37 (29%)	36 (28%)	39 (32%)
Brain, liver or bone	158 (42%)	47 (36%)	59 (46%)	52 (42%)
1L treatments, n (%)				
Chemotherapy (CT)	68 (18%)	41 (32%)	20 (16%)	7 (6%)
1st gen TKI	159 (42%)	83 (64%)	61 (48%)	15 (12%)
2nd gen TKI	53 (14%)	# (~3%)	46 (36%)	<5
3rd gen TKI	99 (26%)	0	0	99 (80%)

If the patient number is less than five (<5), it is displayed as "<5". If the patient number is hidden (#), it means that displaying it would allow the calculation of the exact patient number. This is done to preserve participant anonymity and comply with the General Data Protection Regulation (GDPR).

Treatment patterns (Figure 2)

- 52% (n=83), ~35% and 19% (n=19) of patients receiving 1L 1st gen, 2nd gen, and 3rd gen TKIs respectively died before receiving any 2L treatment. Majority of patients (65%, n=64) treated with 1L 3rd gen TKIs are still on treatment given the recent time when osimertinib became available in Finland (October 2020). This explains why rates of deaths before 2L treatments are different according to treatment categories received in 1L.
- Of those switching from 3rd gen TKIs treatment in 1L and receiving a subsequent therapy (n=15), CT was the most used (n=10, 67%).
- 88% of patients receiving CT in 1L did receive a 2L treatment (n=60), mostly (n=43, 72%) being TKIs.

Figure 2: Treatment line progression from first to second line

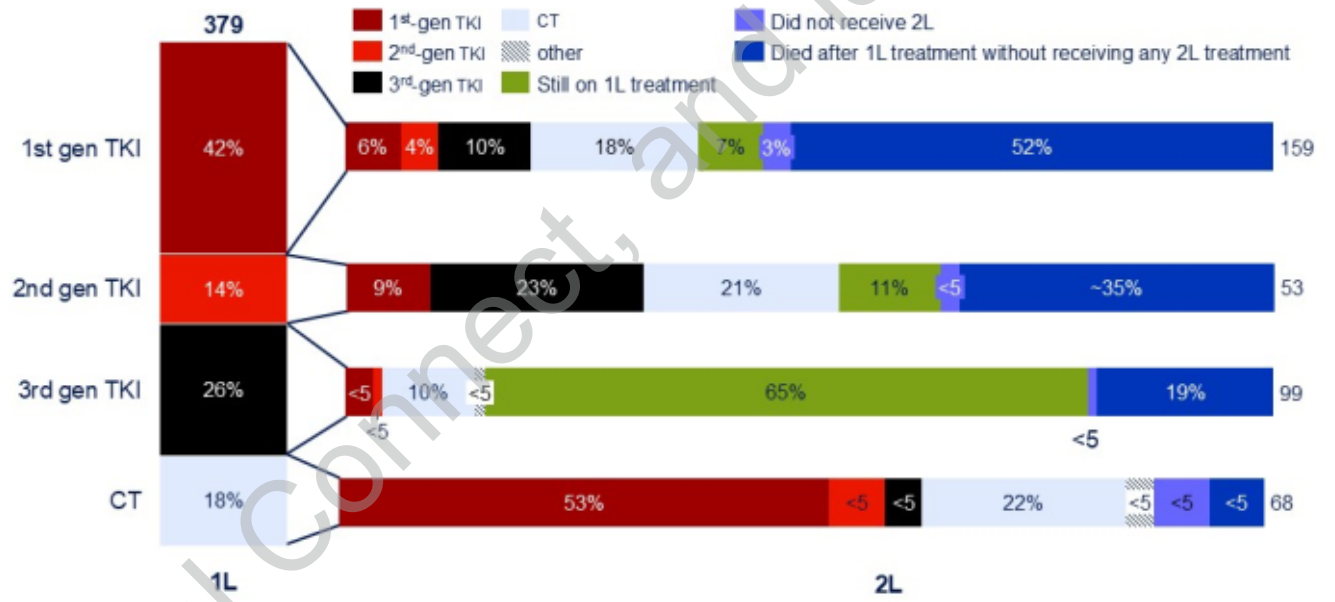
379
1st-gen TKI
2nd-gen TKI
3rd-gen TKI
CT
other
Still on 1L treatment
Did not receive 2L
Died after 1L treatment without receiving any 2L treatment
1st gen TKI
2nd gen TKI
3rd gen TKI
CT
42%
14%
26%
18%
159
53
99
68
1L
2L

Patients in 1L	CT	1st-gen TKI	2nd-gen TKI	3rd-gen TKI	All
Analysis set, patients (n)	68	159	53	99	379
Total number of death events, n (%)	53 (78%)	136 (86%)	40 (75%)	33 (33%)	262 (69%)
Total number of patient alive at the end of follow-up, n (%)	15 (22%)	23 (14%)	13 (25%)	66 (67%)	117 (31%)
Subject disposition					
Subjects who are still on 1L treatment, n (%)	0 (0%)	11 (7%)	#	64 (65%)	81 (21%)
Subjects alive who discontinued 1L treatment and did not receive 2L, n (%)	<5	5 (3%)	<5	<5	11 (3%)
Subjects with 2L treatment, n (%)	60 (88%)	60 (38%)	28 (53%)	15 (15%)	163 (43%)
CT, n	15	28	11	10	65
1st-gen TKI, n	36	10	#	<5	54
2nd-gen TKI, n	<5	6	0	<5	11
3rd-gen TKI, n	<5	16	#	0	31
Other, n	<5	0	0	<5	<5
Subjects who died after 1L treatment without receiving any 2L treatment, n (%)	<5	83 (52%)	# (~35%)	19 (19%)	124 (33%)

Treatment types: 1st-gen TKI, first-generation TKI; 2nd-gen TKI, second generation TKI; 3rd-gen TKI, third-generation TKI, CT, chemotherapy; other, any other NSCLC treatment not classified to previous treatment groups, did not receive 2L, the patients who ended first line treatment but did not receive second line treatment during the study period; died after 1L, the patients who died during the first line treatment of after finishing the first line; still on 1L treatment, patient who did not finish the 1L treatment during the study period. If the patient number is less than five (<5), it is displayed as "<5". If the patient number is hidden (#), it means that displaying it would allow the calculation of the exact patient number. This is done to preserve participant anonymity and comply with the General Data Protection Regulation (GDPR).

TTNT (Figure 3 and Table 2)

- Median TTNT substantially improved over time, from 9.7 months in 2010-2016 to 13.2 months in 2017-2020 to 21.6 months in 2020-2023.
- The TTNT increase from 2010–2016 to 2020–2023 was statistically significant (9.7 vs. 21.6 months; HR: 0.51; 95% CI: 0.37–0.70; p < 0.001).
- While stratifying the results by treatment type received in 1L, median TTNT was longest with 3rd-generation TKI (24.3 months) and shortest with CT (4.3 months).

OS (Figure 4 and Table 3)

- Median OS substantially improved over time, from 19.1 months in 2010-2016 to 23.9 months in 2017-2020 to 29.3 months in 2020-2023.
- The OS increase from 2010–2016 to 2020–2023 was statistically significant (19.1 vs. 29.3 months; HR: 0.66; 95% CI: 0.46–0.94; p=0.021).
- While stratifying the results by treatment type received in 1L, median OS was longest with 3rd-gen TKI (31.1 months) and shortest with 1st-gen TKI (17.7 months).
- The OS outcomes in this RW population were substantially lower than those reported in previous clinical trials[5-6].

Figure 3: Time to next treatment (TTNT) for different time periods

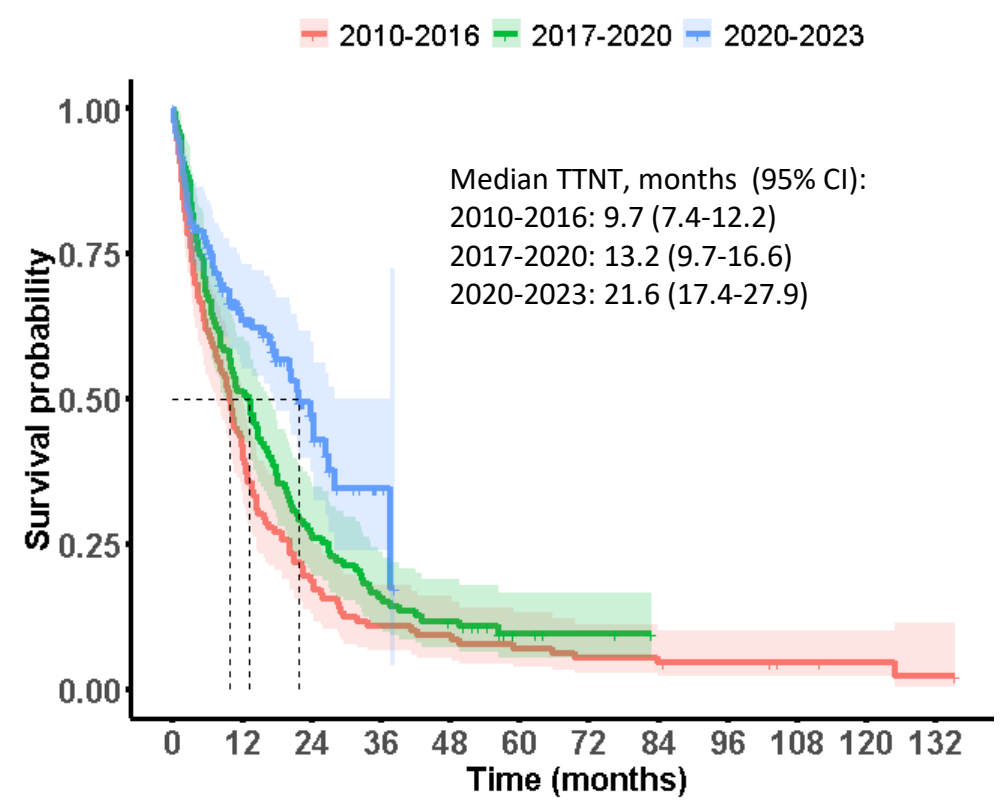
2010-2016
2017-2020
2020-2023
Median TTNT, months (95% CI):
2010-2016: 9.7 (7.4-12.2)
2017-2020: 13.2 (9.7-16.6)
2020-2023: 21.6 (17.4-27.9)
Survival probability
1.00
0.75
0.50
0.25
0.00
0 12 24 36 48 60 72 84 96 108 120 132
Time (months)

Figure 4: Overall survival (OS) for different time periods

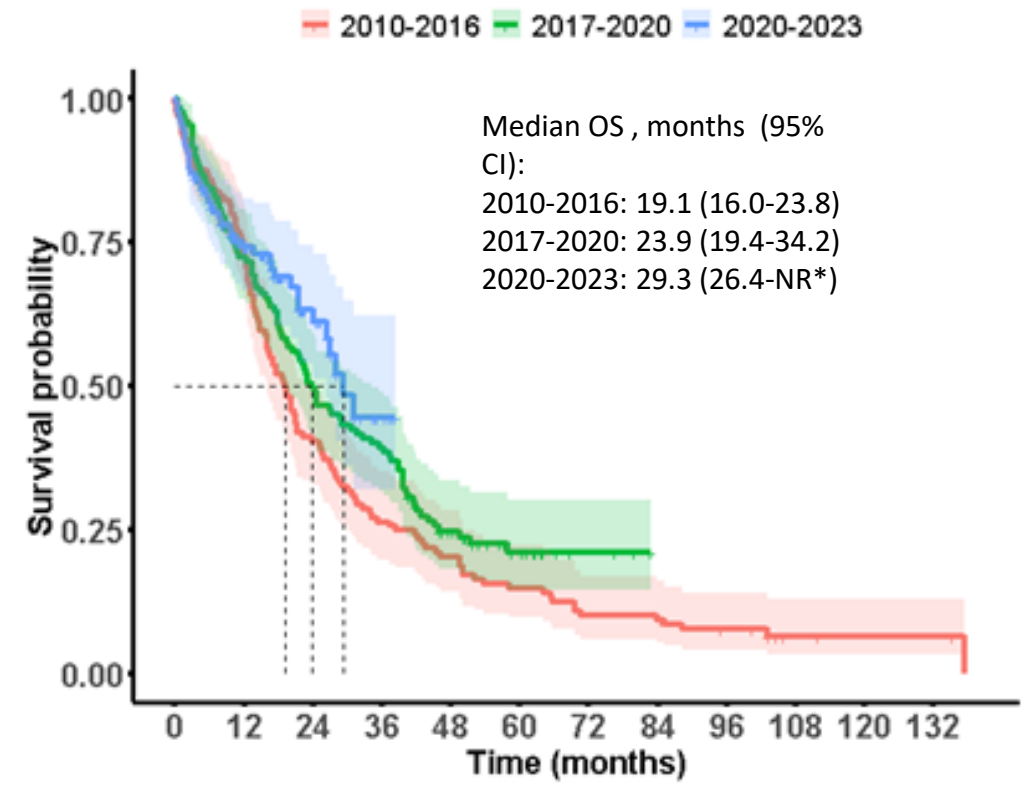
2010-2016
2017-2020
2020-2023
Median OS , months (95% CI):
2010-2016: 19.1 (16.0-23.8)
2017-2020: 23.9 (19.4-34.2)
2020-2023: 29.3 (26.4-NR*)
Survival probability
1.00
0.75
0.50
0.25
0.00
0 12 24 36 48 60 72 84 96 108 120 132
Time (months)

Table 2: Median TTNT for first-line treatment type

1L treatment	Median TTNT (months)	95% CI (months)	Hazard ratio (95% CI, p-value)
1st-gen TKI (n=159)	12.8	10.8-15.7	Ref
2nd-gen TKI (n=53)	14.8	10.2-23.3	0.92 (0.66-1.28, p=0.608)
3rd-gen TKI (n=99)	24.3	20.3-NR*	0.53 (0.38-0.75, p<0.001)
CT (n=68)	4.3	3.32-6.1	2.18 (1.61-2.95, p<0.001)

Table 3: Median OS for first-line treatment type

1L treatment	Median OS (months)	95% CI (months)	Hazard ratio (95% CI, p-value)
1st-gen TKI (n=159)	17.7	14.5-21.2	Ref
2nd-gen TKI (n=53)	26.9	16.6-39.0	0.79 (0.55-1.12, p=0.189)
3rd-gen TKI (n=99)	31.1	26.4-NR*	0.59 (0.40-0.87, p=0.008)
CT (n=68)	25.7	22.9-39.6	0.72 (0.53-0.99, p=0.046)

References

[1]Hirsch, F.R. *et al.* New and emerging targeted treatments in advanced non-small-cell lung cancer. The Lancet. 388(10048):1012-24 (2016) [2]Cheng, Z. *et al* The advance of the third-generation EGFR-TKI in the treatment of non-small cell lung cancer (Review). Oncol Rep. 51(1):16 (2024);. Hendriks ,LE. Oncogene-addicted metastatic non-small-cell lung cancer: ESMO Clinical Practice Guideline for diagnosis, treatment and follow-up. Ann Oncol. 34(4):339-357 (2023). [4]Hsu, W. *et al.* Overview of current systemic management of EGFR-mutant NSCLC. Ann Oncol. 29, i3–i9 (2018);); [5]Ramalingam, SS. *et al.* Overall Survival with Osimertinib in Untreated, EGFR-Mutated Advanced NSCLC. N Engl J Med.382(1):41-50 (2020); [6]Papadimitrakopoulou, VA. *et al.* Osimertinib versus platinum-pemetrexed for patients with EGFR T790M advanced NSCLC and progression on a prior EGFR-tyrosine kinase inhibitor: AURA3 overall survival analysis. Ann Oncol. 31(11) (2020).

Lung Cancer

European Lung Cancer Congress; March 26-29, 2025; Paris, France

This material is distributed for scientific purposes on J&J Medical Connect, and is not for promotional use