58P

Subcutaneous after intravenous amivantamab in advanced NSCLC: Initial results from PALOMA-2

Sun Min Lim[1,*], Ji-Youn Han[2], Jun Zhang[3], Eldsamira Mascarenhas[4], Carlos Henrique Andrade Teixeira[5], Nicolas Girard[6], Nir Peled[7], Susan C Scott[8], Martin Gutierrez[9], Alessandra Bulotta[10], Jiunn Liang Tan[11], Tarek Mekhail[12], Dana-Adriana Botesteanu[13], Ali Alhadab[14], Janine Mahoney[15], Miao Wang[14], Jie Zhang[15], Julia Schuchard[16], Mahadi Baig[13], Flavia Amaral Duarte[17]

*Presenting author: limlove2008@yuhs.ac.

[1]Division of Medical Oncology, Department of Internal Medicine, Yonsei University College of Medicine, Seoul, Republic of Korea; [2]National Cancer Center, Goyang, Republic of Korea; [3]Division of Medical Oncology, Department of Internal Medicine, University of Kansas Medical Center, Kansas City, KS, USA; [4]Instituto D'Or de Pesquisa e Ensino (IDOR), Rio de Janeiro, Brazil; [5]Hospital Alemão Oswaldo Cruz, São Paulo, Brazil; [6]Institut Curie, Institut du Thorax Curie-Montsouris, Paris, France and Paris Saclay University, Université de Versailles Saint-Quentin-en-Yvelines, Paris, France; [7]Shaare Zedek Medical Center, The Hebrew University, Distrito Sul, Israel; [8]The Sidney Kimmel Comprehensive Cancer Center, Johns Hopkins University School of Medicine, Baltimore, MD, USA; [9]Hackensack University Medical Center, Hackensack, NJ, USA; [10]San Raffaele Hospital, Milan, Italy; [11]University of Malaya Medical Center, Kuala Lumpur, Malaysia; [12]AdventHealth Cancer Institute, Orlando, FL, USA; [13]Johnson & Johnson, Raritan, NJ, USA; [14]Johnson & Johnson, San Diego, CA, USA; [15]Johnson & Johnson, Spring House, PA, USA; [16]Johnson & Johnson, Horsham, PA, USA; [17]Personal – Oncologia de Precisão e Personalizada, Belo Horizonte – MG, Brazil

This material is distributed for scientific purposes on J&J Medical Connect, and is not for promotional use

Background

- Amivantamab, an epidermal growth factor receptor (EGFR)–MET bispecific antibody with immune cell–directing activity,[1–3] is approved as an intravenous (IV) formulation in multiple *EGFR*-mutated advanced/metastatic non-small cell lung cancer (NSCLC) settings[4,5]
- In the PALOMA-3 study (ClinicalTrials.gov Identifier: NCT05388669), third-line subcutaneous (SC) amivantamab demonstrated noninferior pharmacokinetics (PK) and objective response rate versus amivantamab IV[6]
- In addition, compared with amivantamab IV, amivantamab SC offered:
 - A 5-fold reduction of infusion-related reactions (13% vs 66%)[6]
 - Substantially faster administration time (4.8 min vs 5.0 h at Cycle 1 Day 1)[6]
 - Higher participant-reported convenience (85% vs 35% at the end of treatment) and reduced medical resource utilization[6,7]
- The phase 2 PALOMA-2 study (ClinicalTrials.gov Identifier: NCT05498428) is a global, parallel-cohort, phase 2 bridging study evaluating the efficacy, safety, and PK of amivantamab-based SC regimens in various *EGFR*-mutant NSCLC settings
- Here we report the initial experience of switching to amivantamab SC after amivantamab IV monotherapy

Methods

- Cohort 4 enrolled participants who previously received amivantamab IV as part of standard of care, an expanded-access program, or rollover from a long-term extension study for ≥8 weeks without dose reduction and evidence of progressive disease (**Figure 1**)
- Administration-related reaction (ARR) was defined per the *Medical Dictionary for Regulatory Activities* preferred term (referred to as infusion-related reactions in prior IV studies)
- Population PK simulations were conducted for amivantamab IV versus SC exposures for the every 2 weeks (Q2W) dose regimen at 3 different dose levels (DL0, DL[-1], DL[-2]) to assess PK comparability
 - Results are based on the re-simulation of the PALOMA-3 study using the final population PK model in which participants received amivantamab IV or SC at DL0, DL(-1), and DL(-2); PK samples were not collected in PALOMA-2 cohort 4
- Participant-reported outcomes (PROs) were assessed using a modified version of the Therapy Administration Satisfaction Questionnaire (mTASQ)
 - The mTASQ is a 12-item questionnaire that measures the impact of treatment mode (SC administration) on physical functioning, psychological functioning, and activities of daily living, convenience, and satisfaction

FIGURE 1: PALOMA-2 study design

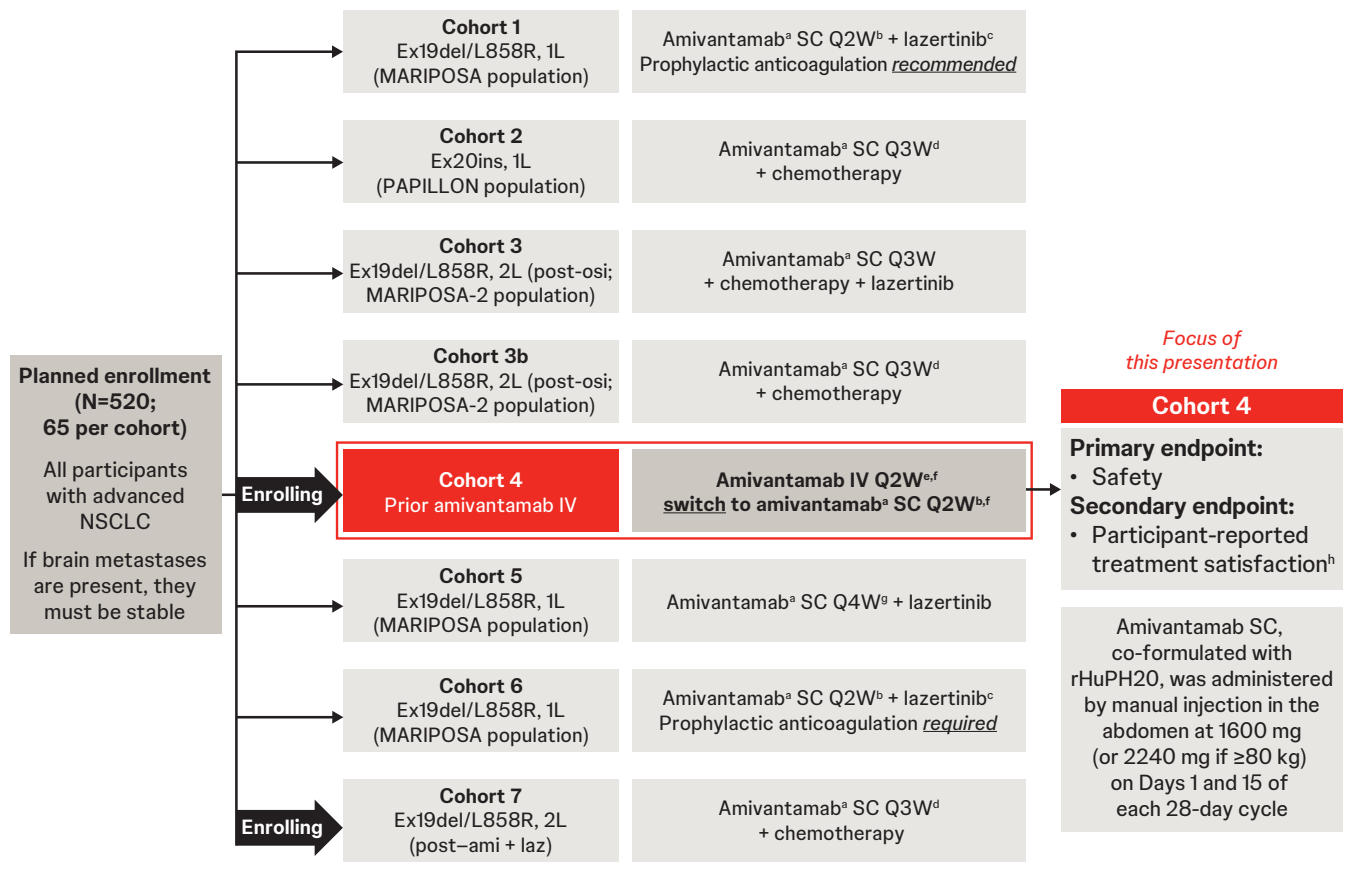
Planned enrollment (N=520; 65 per cohort)
All participants with advanced NSCLC
If brain metastases are present, they must be stable
Enrolling
Cohort 1 Ex19del/L858R, 1L (MARIPOSA population)
Amivantamab[a] SC Q2W[b] + lazertinib[c] Prophylactic anticoagulation recommended
Cohort 2 Ex20ins, 1L (PAPILLON population)
Amivantamab[a] SC Q3W[d] + chemotherapy
Cohort 3 Ex19del/L858R, 2L (post-osi; MARIPOSA-2 population)
Amivantamab[a] SC Q3W + chemotherapy + lazertinib
Cohort 3b Ex19del/L858R, 2L (post-osi; MARIPOSA-2 population)
Amivantamab[a] SC Q3W[d] + chemotherapy
Cohort 4 Prior amivantamab IV
Amivantamab IV Q2W[e,f] switch to amivantamab[a] SC Q2W[b,f]
Cohort 5 Ex19del/L858R, 1L (MARIPOSA population)
Amivantamab[a] SC Q4W[g] + lazertinib
Cohort 6 Ex19del/L858R, 1L (MARIPOSA population)
Amivantamab[a] SC Q2W[b] + lazertinib[c] Prophylactic anticoagulation required
Enrolling
Cohort 7 Ex19del/L858R, 2L (post–ami + laz)
Amivantamab[a] SC Q3W[d] + chemotherapy
Focus of this presentation
Cohort 4
Primary endpoint:
• Safety
Secondary endpoint:
• Participant-reported treatment satisfaction[h]
Amivantamab SC, co-formulated with rHuPH20, was administered by manual injection in the abdomen at 1600 mg (or 2240 mg if ≥80 kg) on Days 1 and 15 of each 28-day cycle

[a]Amivantamab SC was administered by manual injection in the abdomen. [b]Amivantamab SC Q2W dose: 1600 mg (2240 mg if ≥80 kg). [c]Results for Cohorts 1 and 6 were presented previously at the 2024 American Society of Clinical Oncology Annual Meeting.[8] [d]Amivantamab SC Q3W dose: 2400 mg (3360 mg if ≥80 kg). [e]Amivantamab IV Q2W dose: 1050 mg (1400 mg if ≥80 kg). [f]With or without lazertinib. [g]Amivantamab SC Q4W dose: 3520 mg (4640 mg if ≥80 kg). [h]mTASQ was completed by participants at screening (before the IV to SC switch) and following amivantamab SC administration at C1D1, C3D1, and EOT. The original wording of the TASQ specified that the SC injection would take place in the "thigh," but the modified questionnaire specifies that the injection will take place in the "skin."
1L, first-line; 2L, second-line; ami, amivantamab; C, Cycle; D, Day; EGFR, epidermal growth factor receptor; EOT, end of treatment; Ex19del, exon 19 deletion mutation; Ex20ins, exon 20 insertion mutation; IV, intravenous; L858R, exon 21 L858R substitution mutation; laz, lazertinib; mTASQ, modified TASQ; NSCLC, non-small cell lung cancer; osi, osimertinib; Q2W, every 2 weeks; Q3W, every 3 weeks; Q4W, every 4 weeks; rHuPH20, recombinant human hyaluronidase; SC, subcutaneous; TASQ, Therapy Administration Satisfaction Questionnaire.

Results

Baseline demographic and clinical characteristics

- As of October 24, 2024, 26 participants were enrolled in the amivantamab monotherapy cohort (**Table 1**)
 - Among these participants, 25 participants were dosed with amivantamab SC after switching from amivantamab IV
- Median treatment duration was 3.1 months for amivantamab IV and 7.4 months for amivantamab SC
- Median follow-up from first amivantamab SC dose was 9.7 months
- As of the data cutoff, 64% of participants were still ongoing with amivantamab treatment

TABLE 1: Baseline demographic and clinical characteristics

Characteristic	Cohort 4 (n=26)
Median (range) age, years	66 (41–83)
Female, n (%)	15 (58)
History of smoking, n (%)	10 (38)
History of brain metastases, n (%)	8 (31)
Race, n (%)	
Asian	14 (54)
White	10 (38)
Not reported[a]	2 (8)
ECOG PS score, n (%)	
n	25
0	9 (36)
1	16 (64)
EGFR mutation,[b] n (%)	
n	23
L858R	3 (13)
Ex20ins	21 (91)
Adenocarcinoma histology, n (%)	24 (92)

[a]Participant either declined to answer the question or was not able to identify a race. [b]Participants can be included in ≥1 category.
ECOG PS, Eastern Cooperative Oncology Group performance status; EGFR, epidermal growth factor receptor; Ex20ins, exon 20 insertion mutation; L858R, exon 21 L858R substitution mutation.

Safety

- The safety profile of amivantamab SC after switching from amivantamab IV was consistent with that observed in prior studies of amivantamab SC monotherapy,[9] and no new safety signals were identified (**Table 2**)
 - Rash (grouped term inclusive of rash, rash maculo-papular, acne, dermatitis acneiform, rash pustular, and skin lesions) was reported in 10 (40%) participants (grade ≥3, 3 [12%])
 - Only 1 participant discontinued amivantamab SC due to a treatment-related adverse event (interstitial lung disease)
- No ARRs were reported

Participant-reported outcomes

- Most participants (96%) were compliant with mTASQ assessments through Cycle 1
- PROs for amivantamab IV at screening and amivantamab SC at Cycle 1 are shown in **Figure 2**
- By Cycle 1:
 - Most participants were satisfied with amivantamab SC (79%), found it convenient (83%), and preferred it (63%)
 - Among amivantamab SC recipients (n=24), 54% reported feeling unrestricted and 67% reported feeling unbothered by the time for treatment administration compared with 24% and 12% for amivantamab IV (n=25), respectively
- This trend continued or improved further at Cycle 3
- Most participants reported mild or no injection-site symptoms with amivantamab SC at Cycle 1: 71% reported mild or no pain, 83% reported mild or no swelling, and 88% reported mild or no redness
 - Severe injection-site pain was reported by 8% of amivantamab SC recipients (n=24) at Cycle 1, and decreased to none at Cycle 3

Exploratory PK Simulations

- PK met the noninferiority criterion for efficacy when the lower bound of the geometric mean ratio (GMR) 90% confidence interval (CI) for average concentration (C_{avg}) and trough concentration (C_{trough}) was ≥0.8, and for safety when the upper bound of the GMR 90% CI for maximum concentration (C_{max}) was ≤1.25
- Simulated exposures of amivantamab IV versus SC for the Q2W dose regimen were noninferior, which further supports the IV to SC switch at reduced dose levels (**Figure 3**)

TABLE 2: Safety profile of amivantamab SC monotherapy

Most common treatment-emergent AEs (≥10%), n (%)	Cohort 4[a] (n=25)[b] All grades	Cohort 4[a] (n=25)[b] Grade ≥3
Associated with EGFR inhibition		
Paronychia	11 (44)	1 (4)
Rash[c]	5 (20)	0
Stomatitis	4 (16)	0
Pruritus	3 (12)	0
Associated with MET inhibition		
Hypoalbuminemia	10 (40)	1 (4)
Peripheral edema	4 (16)	0
Other		
Dyspnea	6 (24)	1 (4)
Aspartate aminotransferase increased	6 (24)	0
Hypocalcemia	5 (20)	0
Alanine aminotransferase increased	5 (20)	0
Asthenia	4 (16)	1 (4)
Decreased appetite	4 (16)	0
Neutropenia	4 (16)	0
Edema	4 (16)	0
Pneumonia	3 (12)	3 (12)
Fatigue	3 (12)	0
Pyrexia	3 (12)	0
Epistaxis	3 (12)	0
Dry eye	3 (12)	0
Localized edema	3 (12)	0

[a]Safety analysis set, defined as all participants who switched from amivantamab IV and received an amivantamab SC dose. [b]One participant received their first dose of amivantamab SC after the data cutoff date. [c]Preferred term.
AE, adverse event; EGFR, epidermal growth factor receptor; IV, intravenous; SC, subcutaneous.

FIGURE 2: Participants reporting mTASQ assessments

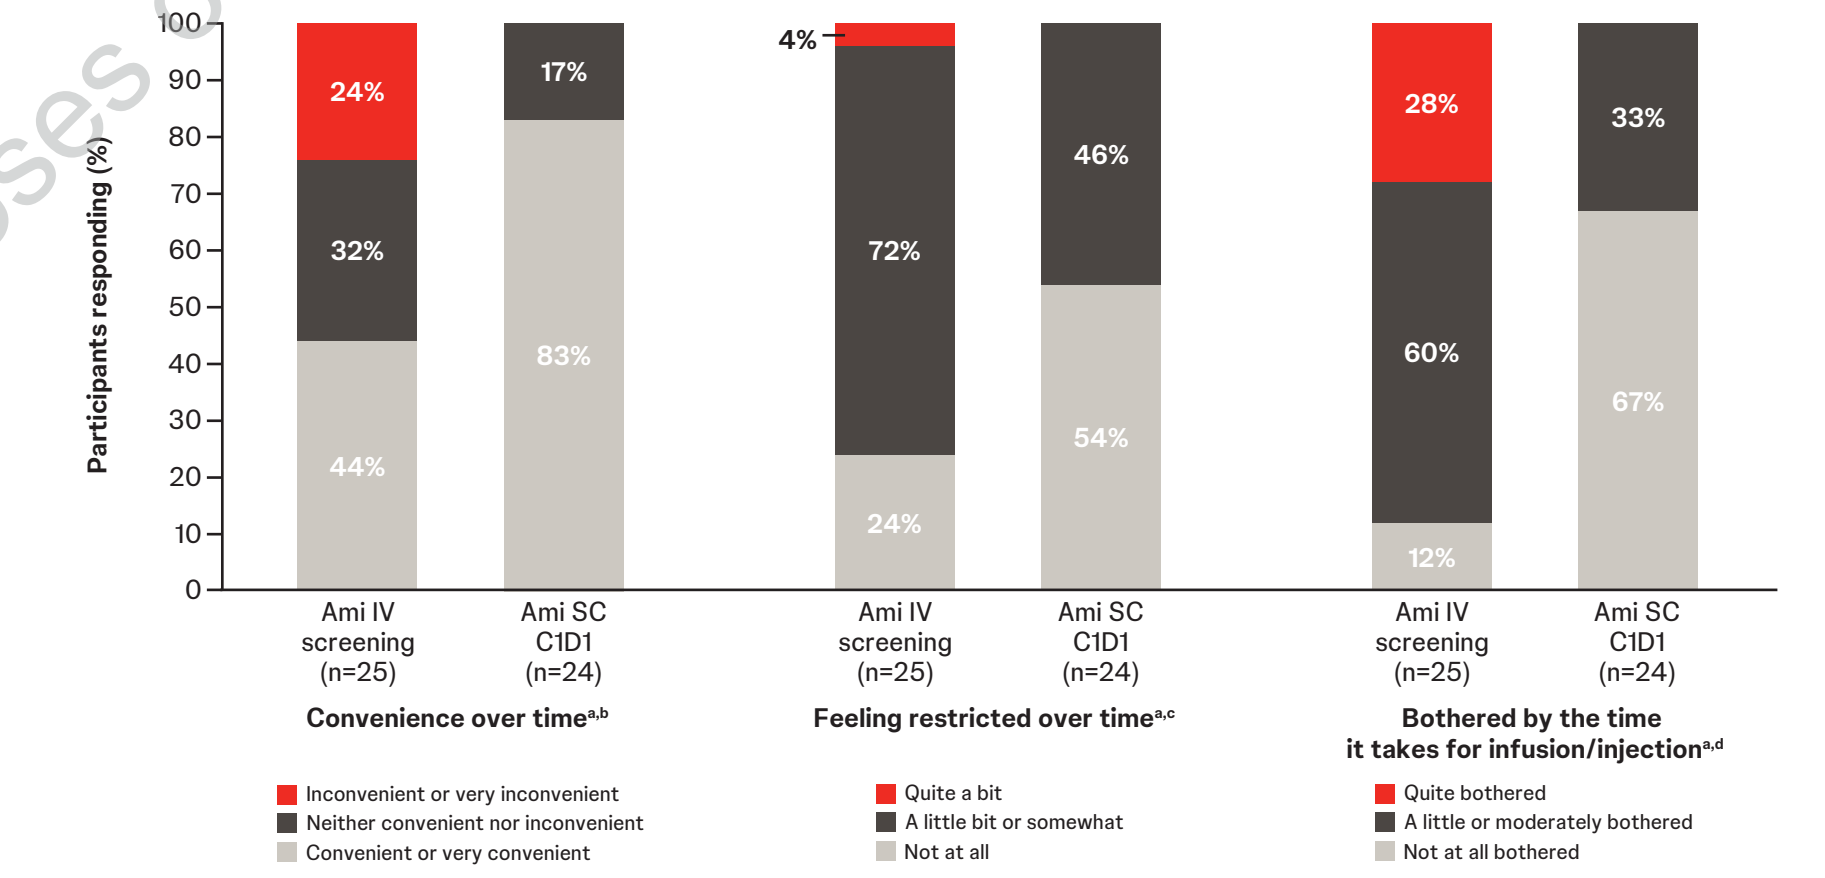
Participants responding (%)
100
90
80
70
60
50
40
30
20
10
0
24%
32%
44%
17%
83%
4%
72%
24%
46%
54%
28%
60%
12%
33%
67%
Ami IV screening (n=25)
Ami SC C1D1 (n=24)
Ami IV screening (n=25)
Ami SC C1D1 (n=24)
Ami IV screening (n=25)
Ami SC C1D1 (n=24)
Convenience over time[a,b]
Feeling restricted over time[a,c]
Bothered by the time it takes for infusion/injection[a,d]
Inconvenient or very inconvenient
Neither convenient nor inconvenient
Convenient or very convenient
Quite a bit
A little bit or somewhat
Not at all
Quite bothered
A little or moderately bothered
Not at all bothered

[a]The mTASQ for IV injection was completed at screening. [b]Based on participant responses to mTASQ Item #6, "How convenient is it for you to get your SC injection?" [c]Based on participant responses to mTASQ Item #5, "When receiving the SC injection, how restricted did you feel?" [d]Based on participant responses to mTASQ Item #7, "How bothered are you by the amount of time it takes to have the SC injection?"
Ami, amivantamab; C, Cycle; D, Day; IV, intravenous; mTASQ, modified Therapy Administration Satisfaction Questionnaire; SC, subcutaneous.

FIGURE 3: PK simulations for the Q2W regimen[a]

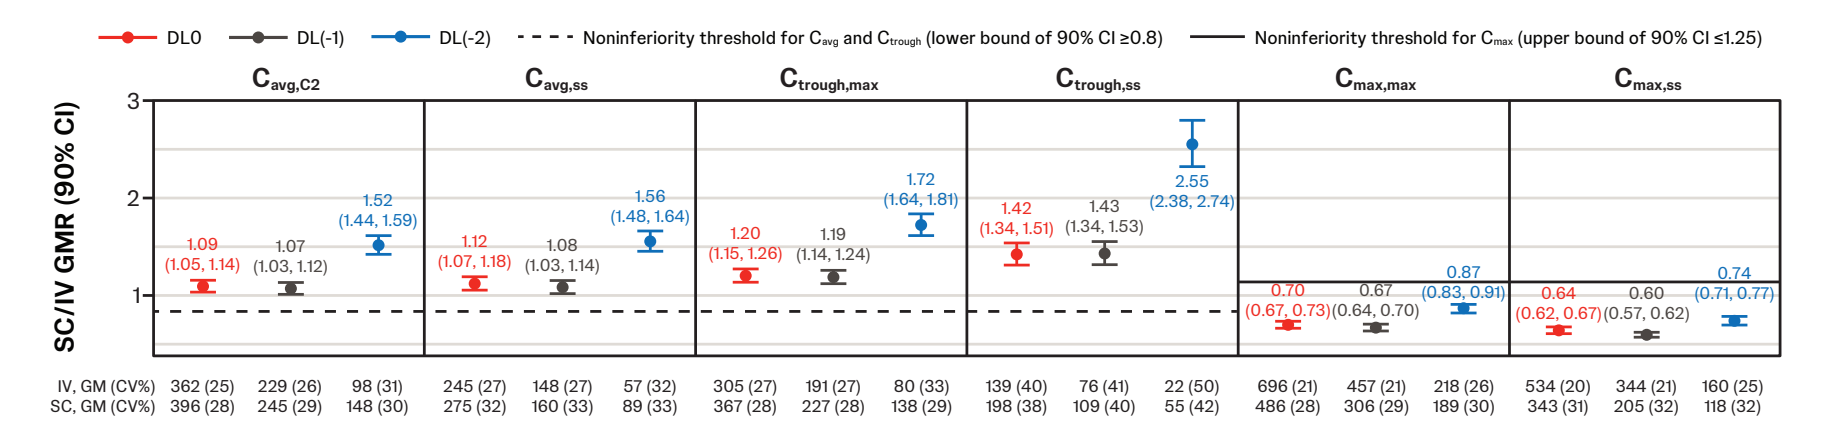
DL0
DL(-1)
DL(-2)
Noninferiority threshold for Cavg and Ctrough (lower bound of 90% CI ≥0.8)
Noninferiority threshold for Cmax (upper bound of 90% CI ≤1.25)
SC/IV GMR (90% CI)
Cavg,C2
Cavg,ss
Ctrough,max
Ctrough,ss
Cmax,max
Cmax,ss
1.09 (1.05, 1.14)
1.07 (1.03, 1.12)
1.52 (1.44, 1.59)
1.12 (1.07, 1.18)
1.08 (1.03, 1.14)
1.56 (1.48, 1.64)
1.20 (1.15, 1.26)
1.19 (1.14, 1.24)
1.72 (1.64, 1.81)
1.42 (1.34, 1.51)
1.43 (1.34, 1.53)
2.55 (2.38, 2.74)
0.70 (0.67, 0.73)
0.67 (0.64, 0.70)
0.87 (0.83, 0.91)
0.64 (0.62, 0.67)
0.60 (0.57, 0.62)
0.74 (0.71, 0.77)
IV, GM (CV%) 362 (25) 229 (26) 98 (31) 245 (27) 148 (27) 57 (32) 305 (27) 191 (27) 80 (33) 139 (40) 76 (41) 22 (50) 696 (21) 457 (21) 218 (26) 534 (20) 344 (21) 160 (25)
SC, GM (CV%) 396 (28) 245 (29) 148 (30) 275 (32) 160 (33) 89 (33) 367 (28) 227 (28) 138 (29) 198 (38) 109 (40) 55 (42) 486 (28) 306 (29) 189 (30) 343 (31) 205 (32) 118 (32)

Note: PK noninferiority criteria to ensure comparable efficacy (lower bound of the GMR 90% CI ≥0.8 for C_{avg} and C_{trough}) and safety (upper bound of the GMR 90% CI ≤1.25 for C_{max}) were met for all DLs.
[a]IV: DL0, 1050 mg (1400 mg if ≥80 kg); DL(-1), 700 mg (1050 mg if ≥80 kg); DL(-2), 350 mg (700 mg if ≥80 kg); SC: DL0, 1600 mg (2240 mg if ≥80 kg); DL(-1), 1050 mg (1600 mg if ≥80 kg); DL(-2), 700 mg (1050 mg if ≥80 kg).
C, Cycle; C_{avg}, average concentration; C_{max}, maximum concentration; C_{trough}, trough concentration; CI, confidence interval; CV, coefficient of variation; DL, dose level; GM, geometric mean; GMR, geometric mean ratio; IV, intravenous; max, maximum; PK, pharmacokinetics; Q2W, every 2 weeks; SC, subcutaneous; SS, steady state.

Key takeaways

Switching from intravenous amivantamab to subcutaneous (SC) amivantamab monotherapy is feasible and safe, with no administration-related reactions reported among participants with epidermal growth factor receptor (*EGFR*)-mutated non-small cell lung cancer

The SC administration of amivantamab is convenient and preferred by participants

Conclusions

The safety profile of participants who switched to amivantamab SC from amivantamab IV was similar to the safety profile previously observed with amivantamab SC monotherapy,[9] demonstrating that the IV to SC switch can occur safely

Most participants were satisfied with amivantamab SC, found it convenient, and preferred it over prior amivantamab IV

Acknowledgments

We would like to acknowledge the individuals who participated in these studies and their families and caregivers, the physicians and nurses who cared for participants and the staff members who supported these clinical trials, and the staff members at the study sites and those who were involved in data collection/analyses. Medical writing assistance was provided by Lumanity Communications Inc. and funded by Johnson & Johnson.

Disclosures

SML received research funding from AstraZeneca, BeiGene, Bristol Myers Squibb, Boehringer Ingelheim, BridgeBio, Daiichi Sankyo, Gilead, GSK, Jiangsu Hengrui Pharmaceuticals, J INTS BIO, Oscotec, Roche, Yuhan Corporation, and Johnson & Johnson; and participated in advisory board meetings for AstraZeneca, Boehringer Ingelheim, Bristol Myers Squibb, Eli Lilly, Janssen, Takeda, Daiichi Sankyo, Yuhan Corporation, Guardant Health, Amgen, J INTS BIO, Therapex, and Johnson & Johnson. **J-YH** received consulting fees from AstraZeneca, AbbVie, LG Chem, Daewoong Pharmaceutical, Lantern Pharma, Janssen, Takeda, Roche, Amgen, Daiichi Sankyo, Oncovix, Novartis, Bristol Myers Squibb, Merck, and Pfizer; received honoraria from AstraZeneca, Takeda, Novartis, Pfizer, Janssen, Merck, Yuhan Corporation, and Roche; received payment for expert testimony from AstraZeneca; and participated in advisory board meetings for Janssen and AstraZeneca. **JZ** received grants or contracts from AbbVie, AstraZeneca, BeiGene, BridgeBio, Genentech, Jiangsu Hengrui Pharmaceuticals, InnoCare Pharma, Janssen, KAHR, Eli Lilly, Merck, Mirati Therapeutics, Bristol Myers Squibb, Nilogen Oncosystems, and Novartis; received consulting fees from AstraZeneca, Bristol Myers Squibb, Fosun International, Jiangsu Hengrui Pharmaceuticals, Eli Lilly, Mirati Therapeutics, Novocure, and Sanofi; received honoraria from AstraZeneca, MJH Life Sciences, Novartis, Regeneron, and Sanofi; planned patents with application serial no. 63/705,859; and participated in advisory board meetings for Jiangsu Hengrui Pharmaceuticals. **EM** received honoraria from Janssen, AstraZeneca, Roche, Pfizer, Merck Sharp & Dohme, and Daiichi Sankyo; received support for attending meetings and/or travel from Roche and Merck Sharp & Dohme; and participated in advisory board meetings for Pfizer, AstraZeneca, and Roche. **CHAT** received honoraria from Janssen, Merck Sharp & Dohme, AstraZeneca, and Roche; received support for attending meetings and/or travel from Daiichi Sankyo, Janssen, and Merck Sharp & Dohme; and participated in advisory board meetings for Merck Sharp & Dohme, AstraZeneca, and Eli Lilly. **NG** received consulting fees from Roche, Eli Lilly, AstraZeneca, Novartis, Pfizer, Bristol Myers Squibb, Merck Sharp & Dohme, Takeda, Janssen, Sanofi, Amgen, Gilead, BeiGene, AbbVie, Daiichi Sankyo/AstraZeneca, LEO Pharma, and Ipsen; received travel, accommodations, or expenses paid by Roche; received research funding from Roche, AstraZeneca, Bristol Myers Squibb, and Merck Sharp & Dohme Avenir; and has a family member employed by AstraZeneca. **NP** received honoraria and consulting fees from Johnson & Johnson. **SCS** received research funding from Mirati Therapeutics, Bristol Myers Squibb, and Janssen; received consulting fees from Amgen, AstraZeneca, Foundation Medicine, Genentech/Roche, Regeneron, and Tempus. **MG** received consulting fees from Sanofi and owns stock or stock options in Cota and Owens & Minor, Inc. **AB** received honoraria from Bristol Myers Squibb, Merck Sharp & Dohme, AstraZeneca, and Eli Lilly; and participated in advisory board meetings for AstraZeneca, Bristol Myers Squibb, and Roche. **JLT** received honoraria from Pfizer, AstraZeneca, and Merck Sharp & Dohme; and received consulting fees from Johnson & Johnson and Bristol Myers Squibb. **D-AB**, **AA**, **JM**, **MW**, **JZ**, **JS**, and **MB** are employees of and may hold stock in Johnson & Johnson. **FAD** received honoraria from Roche, Merck Sharp & Dohme, Bristol Myers Squibb, Eli Lilly, Novartis, Johnson & Johnson, and AstraZeneca; received payment for expert testimony from Takeda, Johnson & Johnson, Daiichi Sankyo, and Pfizer; and received support for attending meetings and/or travel from Takeda, Johnson & Johnson, Daiichi Sankyo, Pfizer, and AstraZeneca. **TM** has nothing to disclose.

Lung Cancer

Scan the QR code

https://www.congresshub.com/Oncology/ELCC2025/Amivantamab/Lim
Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Presented by SM Lim at the European Lung Cancer Congress (ELCC); March 26–29, 2025; Paris, France.

REFERENCES:

1. Moores SL, et al. *Cancer Res*. 2016;76(13):3942–3953. 2. Vijayaraghavan S, et al. *Mol Cancer Ther*. 2020;19(10):2044–2056. 3. Yun J, et al. *Cancer Discov*. 2020;10(8):1194–1209. 4. RYBREVANT® (amivantamab-vmjw) injection, for intravenous use [package insert]. Janssen Biotech, Inc.; 2025. 5. RYBREVANT® (amivantamab-vmjw). Product information. Janssen Biologics B.V.; 2025. 6. Leighl NB, et al. *J Clin Oncol*. 2024;42(30):3593–3605. 7. Alexander M, et al. Presented at: World Conference on Lung Cancer (WCLC) Annual Meeting; September 7–10, 2024; San Diego, CA, USA. 8. Scott SC, et al. Presented at: American Society for Clinical Oncology (ASCO) Annual Meeting; May 31–June 4, 2024; Chicago, IL, USA. 9. Minchom AR, et al. *J Clin Oncol*. 2023;41(16 suppl):9126.